Early stage Eye-fixations reveal Belief-driven Bias in Correlation Perception

Minsuk Chang[1], Adam Malitek[2], Keisuke Fukuda[2,3], Cindy Xiong Bearfield[1]

1 - Georgia Institute of Technology
2 - University of Toronto Mississauga
3 - University of Toronto

Try Our Demo!

Background & Motivation

- Prior beliefs can bias humans' chart perception, even in objective tasks like estimating scatterplot correlations.
- This phenomenon, known as motivated perception, poses challenges for accurately interpreting data visualizations.
- We aim to use **gaze pattern** as a proxy to estimate viewers' belief and bias in scatterplot correlation estimation task.

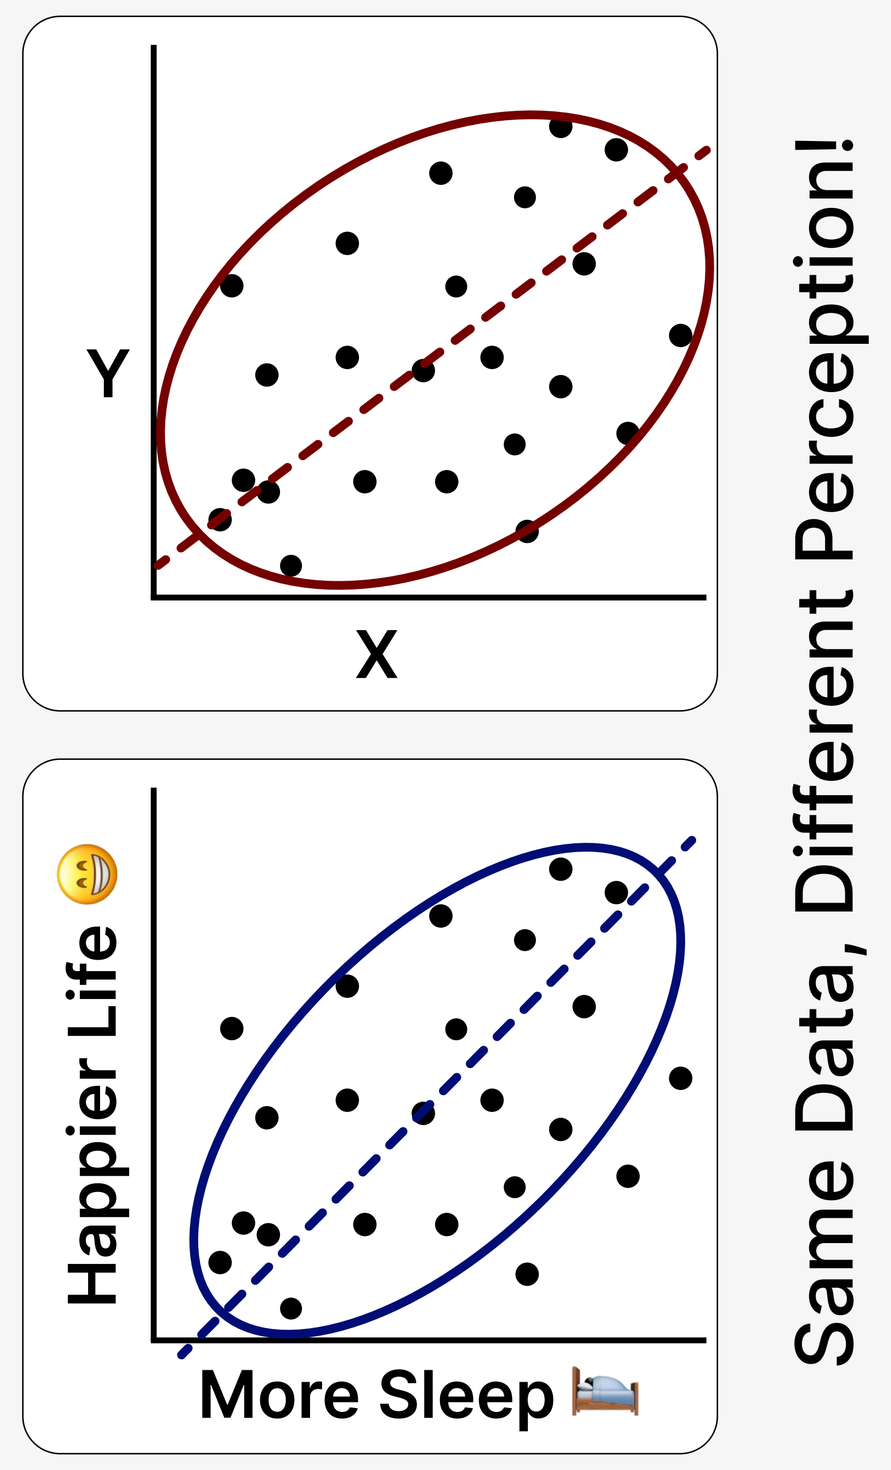
Y
X
Happier Life
More Sleep
Same Data, Different Perception!

Task Design

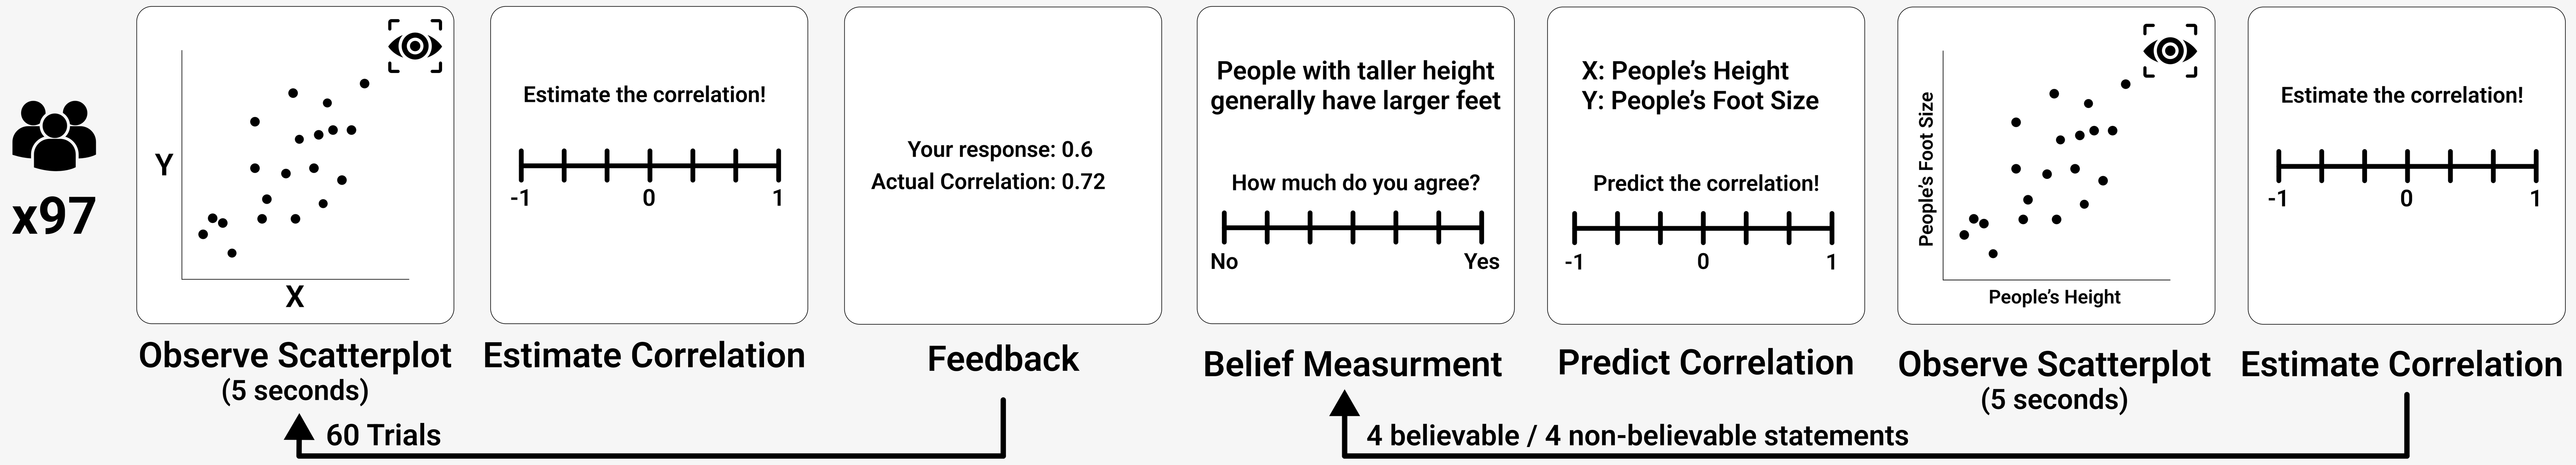
Phase 1 - Calibration
Phase 2 - Belief-Modulated Perception
x97
Y
X
Estimate the correlation!
-1
0
1
Your response: 0.6
Actual Correlation: 0.72
People with taller height generally have larger feet
How much do you agree?
No
Yes
X: People's Height
Y: People's Foot Size
Predict the correlation!
People's Foot Size
People's Height
Observe Scatterplot
(5 seconds)
Estimate Correlation
Feedback
Belief Measurment
Predict Correlation
Observe Scatterplot
(5 seconds)
Estimate Correlation
60 Trials
4 believable / 4 non-believable statements

Result 1 - Level of Prior Belief

Baseline beliefs of different statements

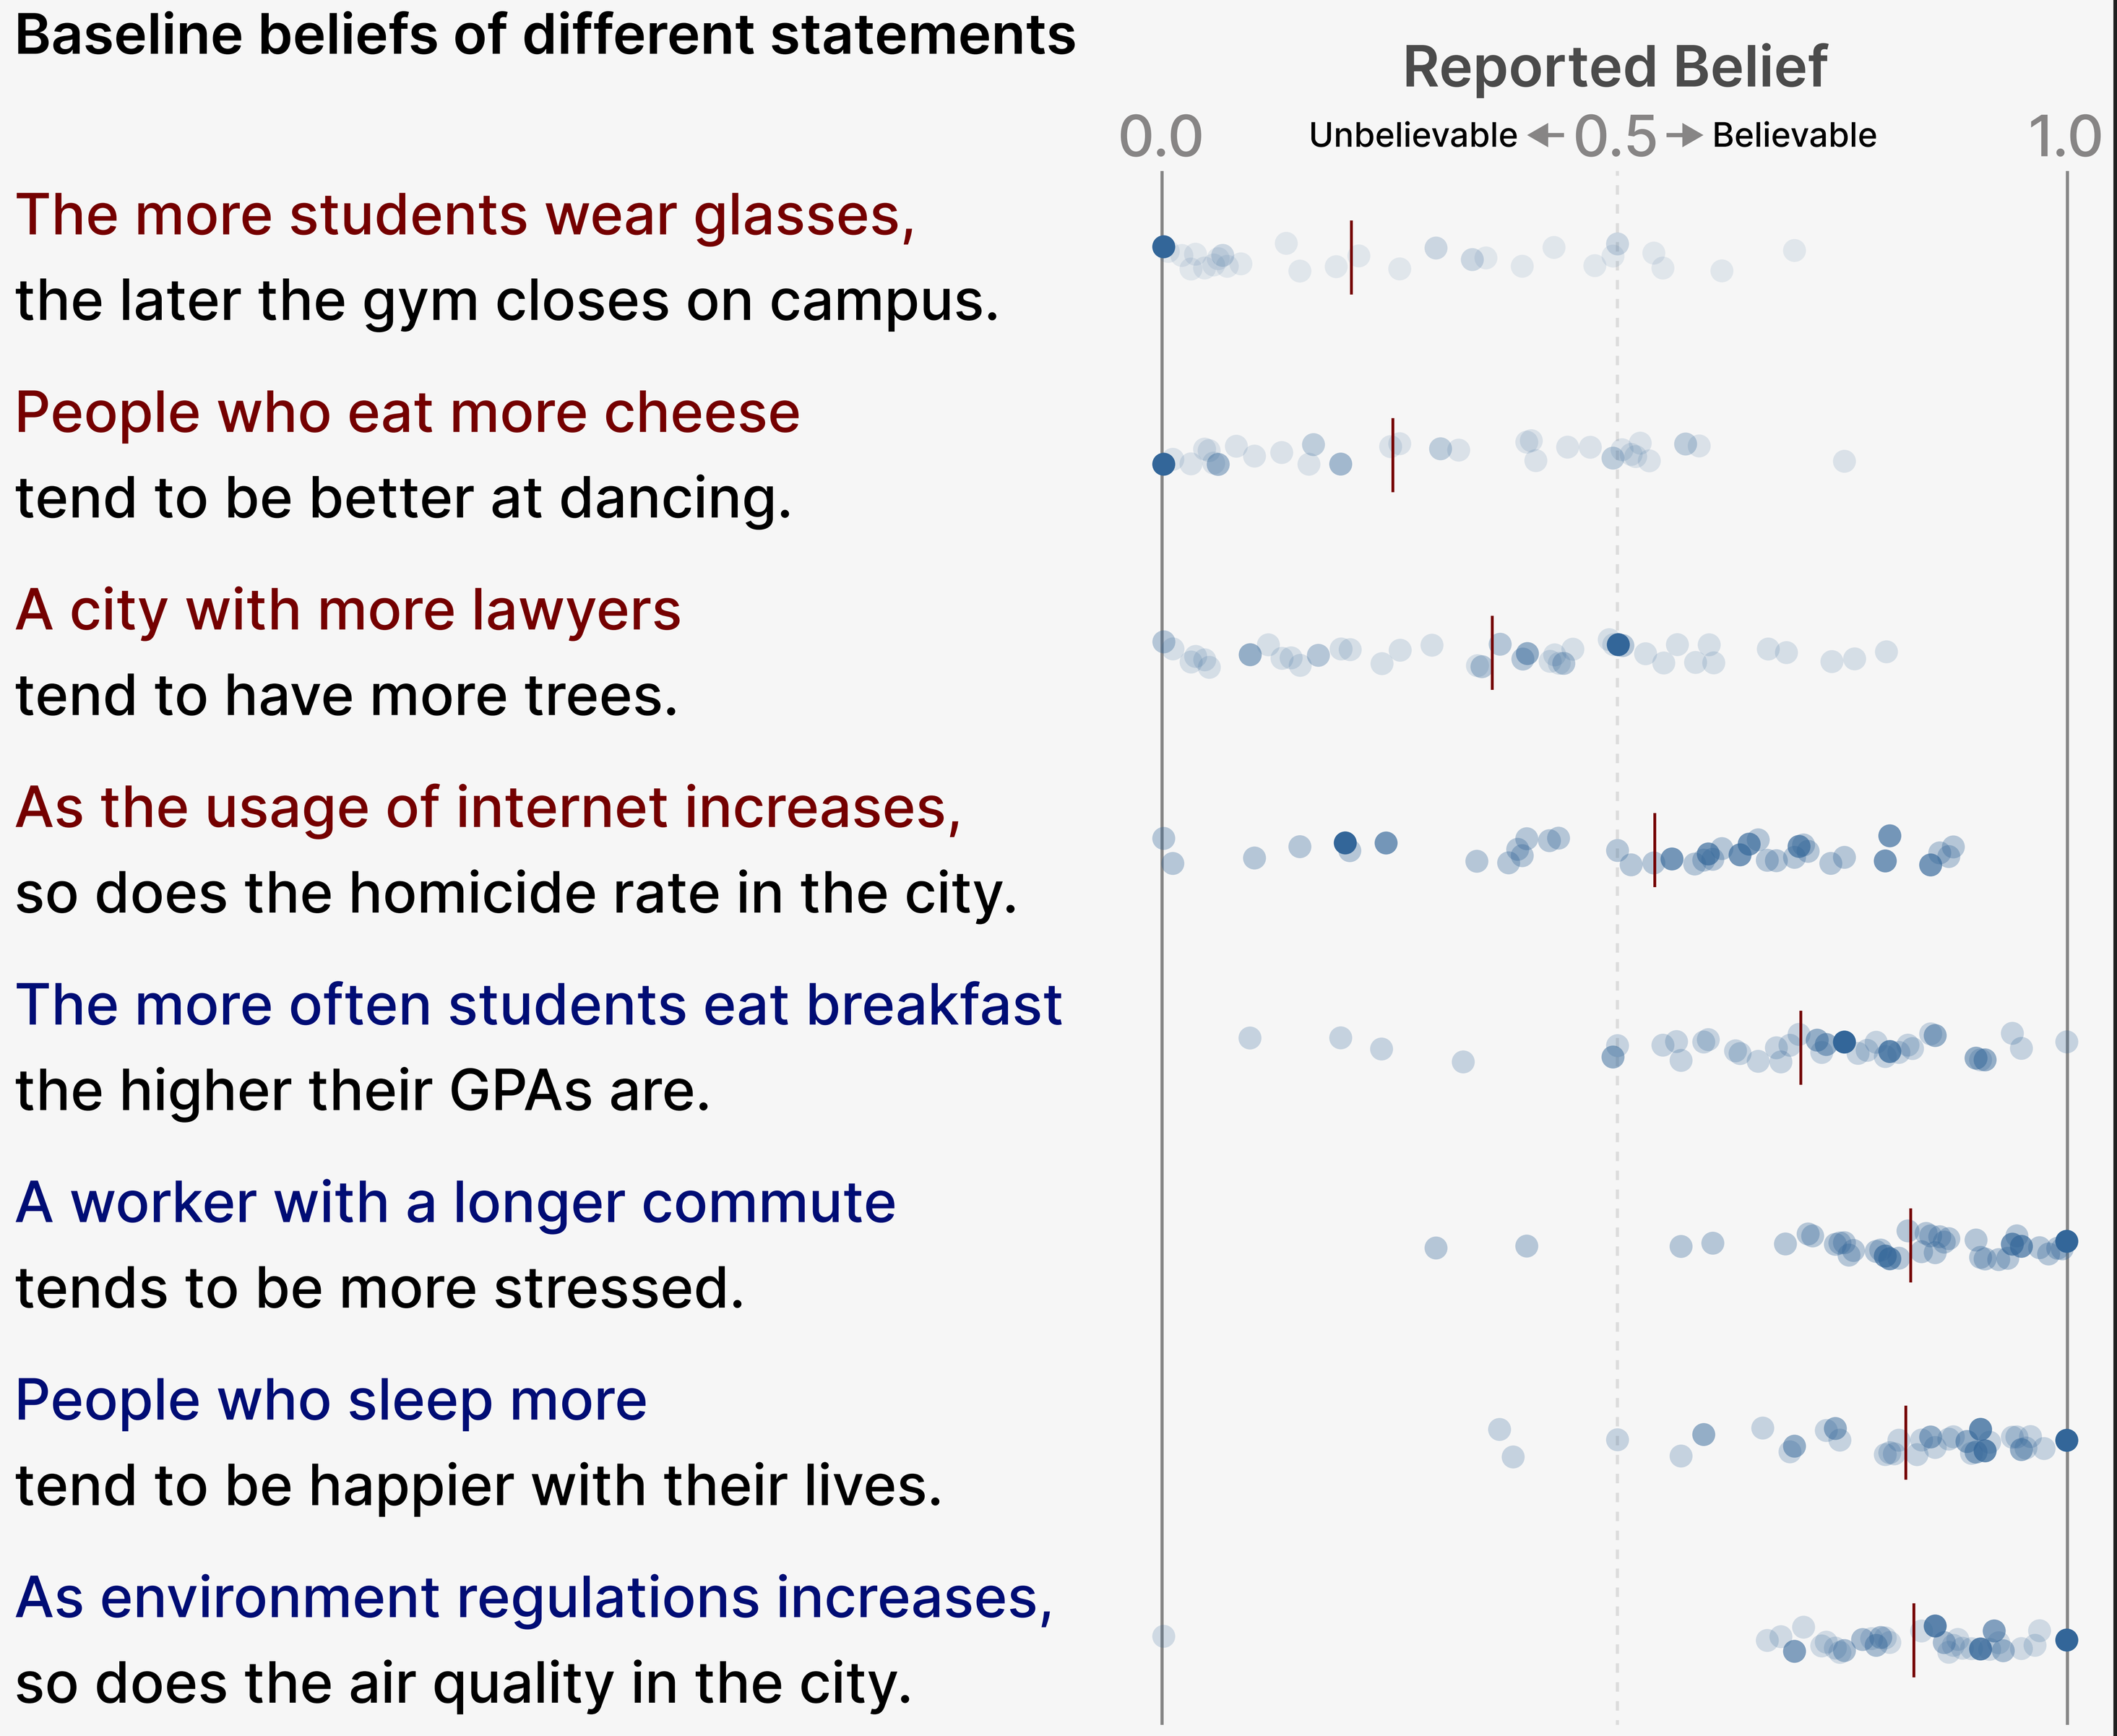
Reported Belief
0.0
Unbelievable
0.5
Believable
1.0
The more students wear glasses,
the later the gym closes on campus.
People who eat more cheese
tend to be better at dancing.
A city with more lawyers
tend to have more trees.
As the usage of internet increases,
so does the homicide rate in the city.
The more often students eat breakfast
the higher their GPAs are.
A worker with a longer commute
tends to be more stressed.
People who sleep more
tend to be happier with their lives.
As environment regulations increases,
so does the air quality in the city.

People hold **diverse prior beliefs** about the same statement

→ Perception of plausibility are inherently **subjective**

Result 2 - Analyzing Gaze Dynamics

We analyzed how gaze patterns evolve over time by calculating the **correlation of fixation points**.

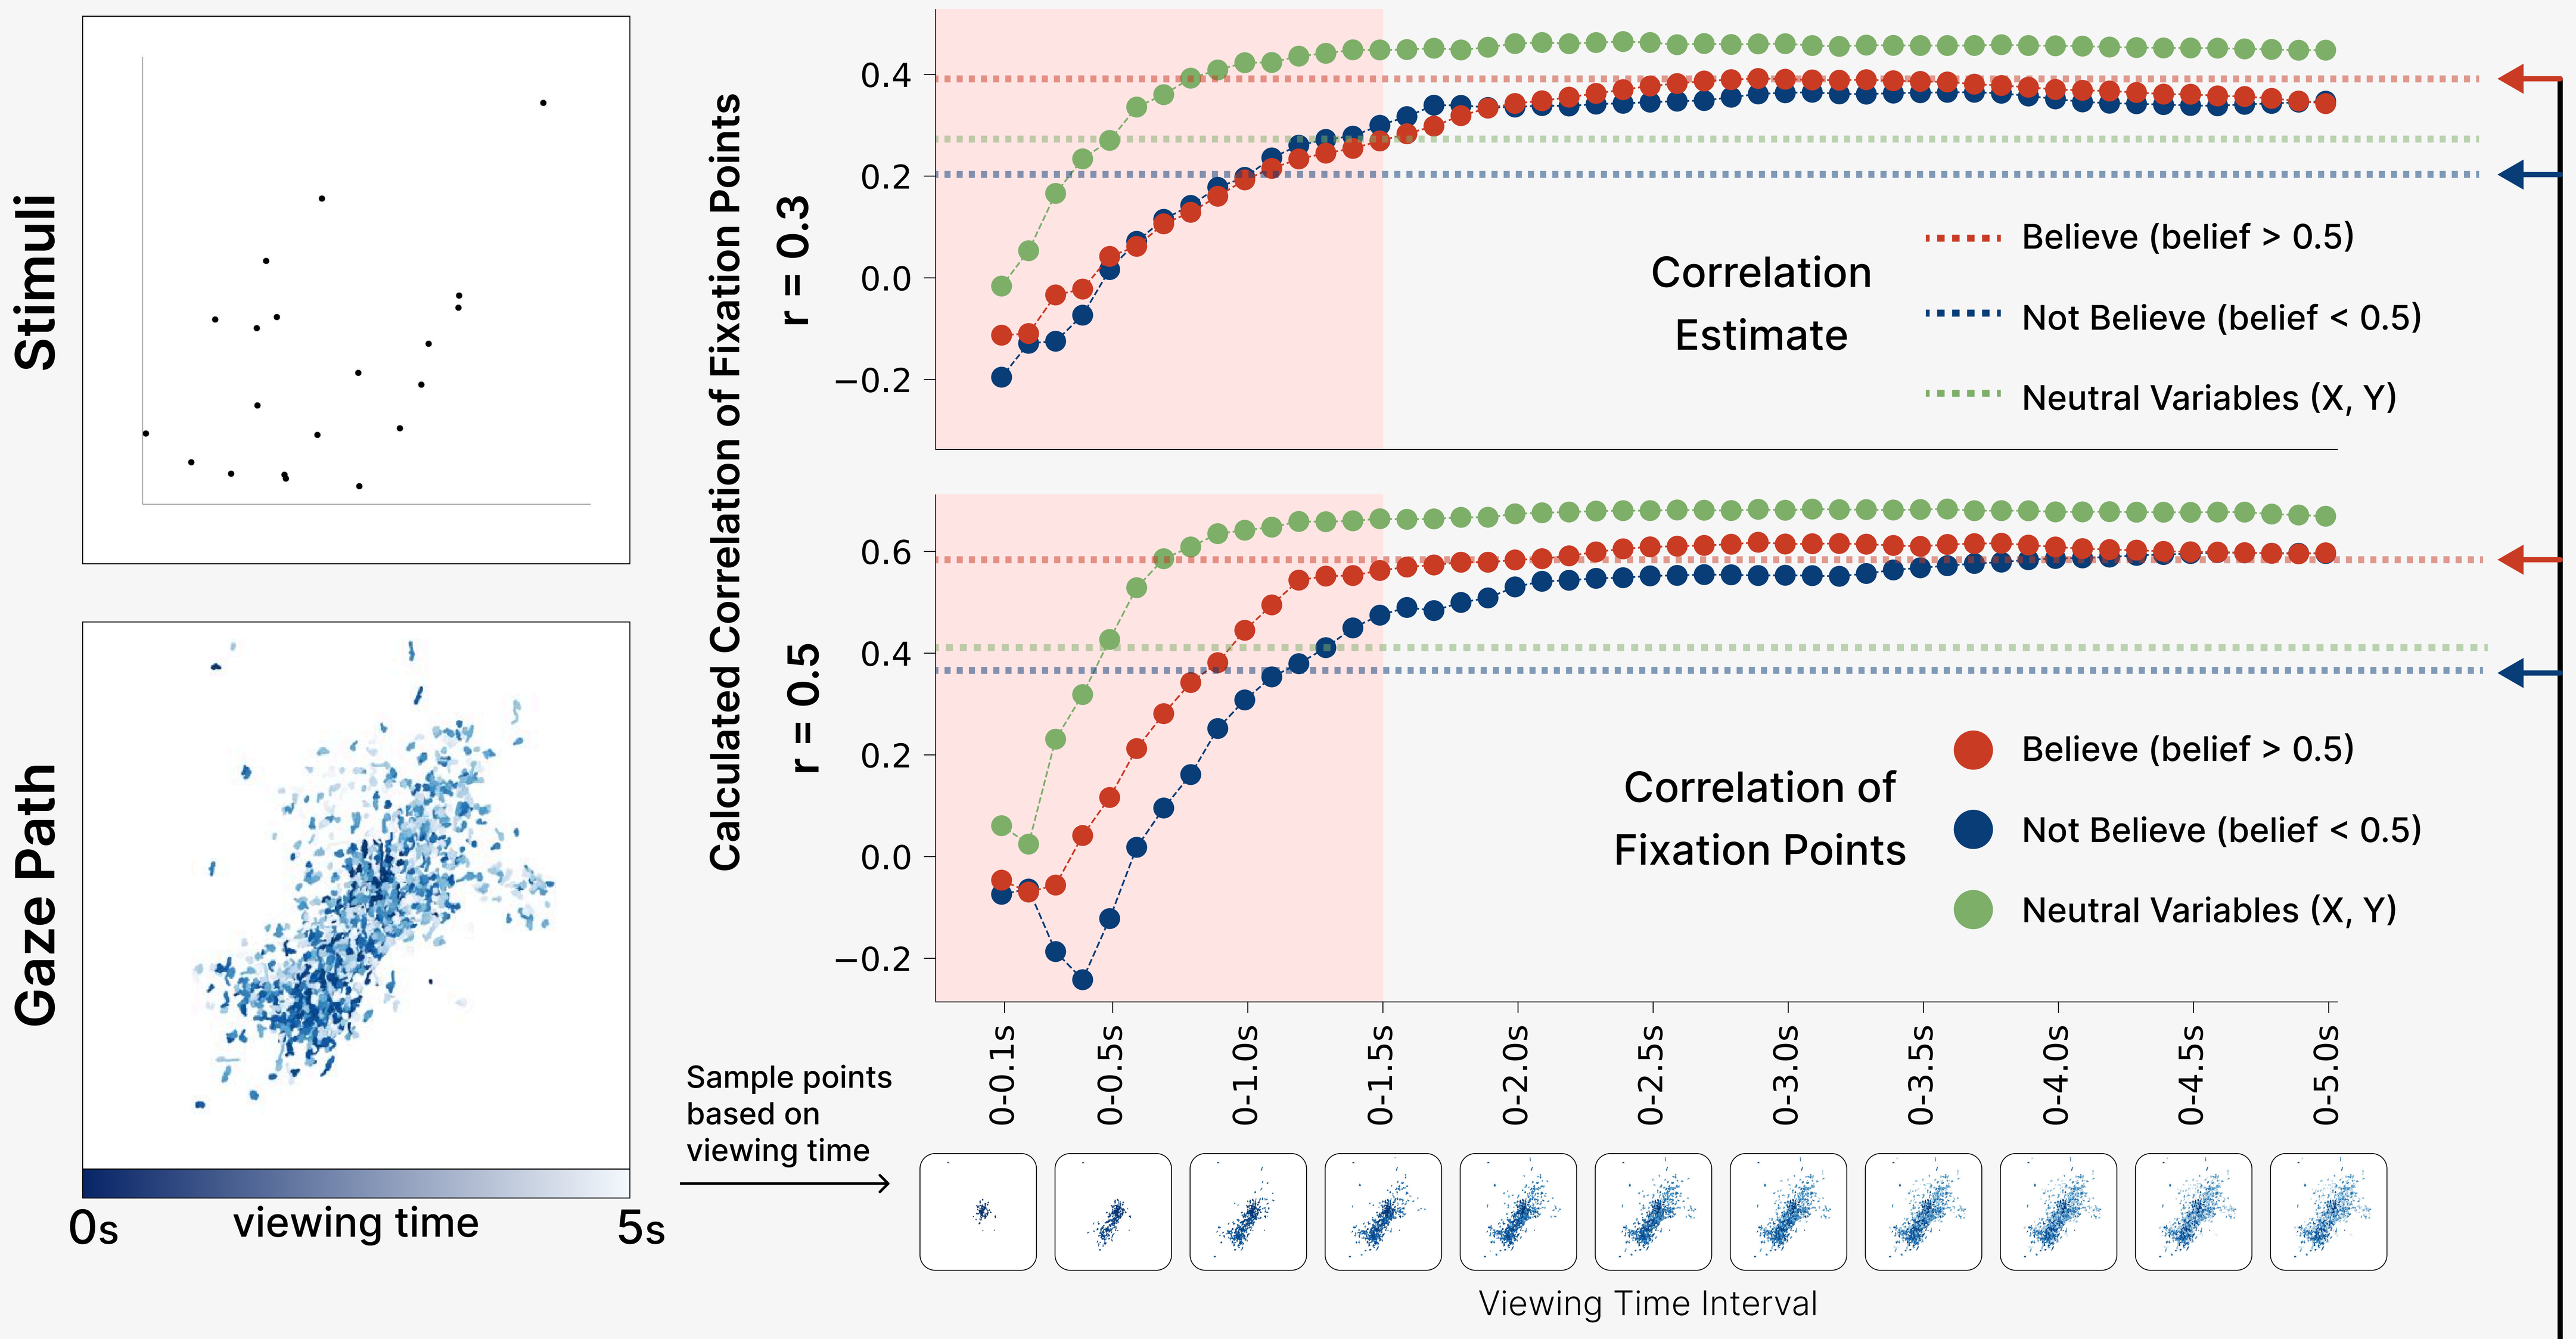
Stimuli
Gaze Path
0s
viewing time
5s
Calculated Correlation of Fixation Points
r = 0.3
r = 0.5
0.4
0.2
0.0
−0.2
0.6
Correlation Estimate
Believe (belief > 0.5)
Not Believe (belief < 0.5)
Neutral Variables (X, Y)
Correlation of Fixation Points
Sample points based on viewing time
0-0.1s
0-0.5s
0-1.0s
0-1.5s
0-2.0s
0-2.5s
0-3.0s
0-3.5s
0-4.0s
0-4.5s
0-5.0s
Viewing Time Interval

When people **believe**/**not believe** the statement, their estimation of the correlation **over/undershoot**

Believable & Non-believable & Neutral condition's convergence speed is different!

Result 3 - AI-driven Analysis

Deep learning model that predicts belief with 1 second fixation points

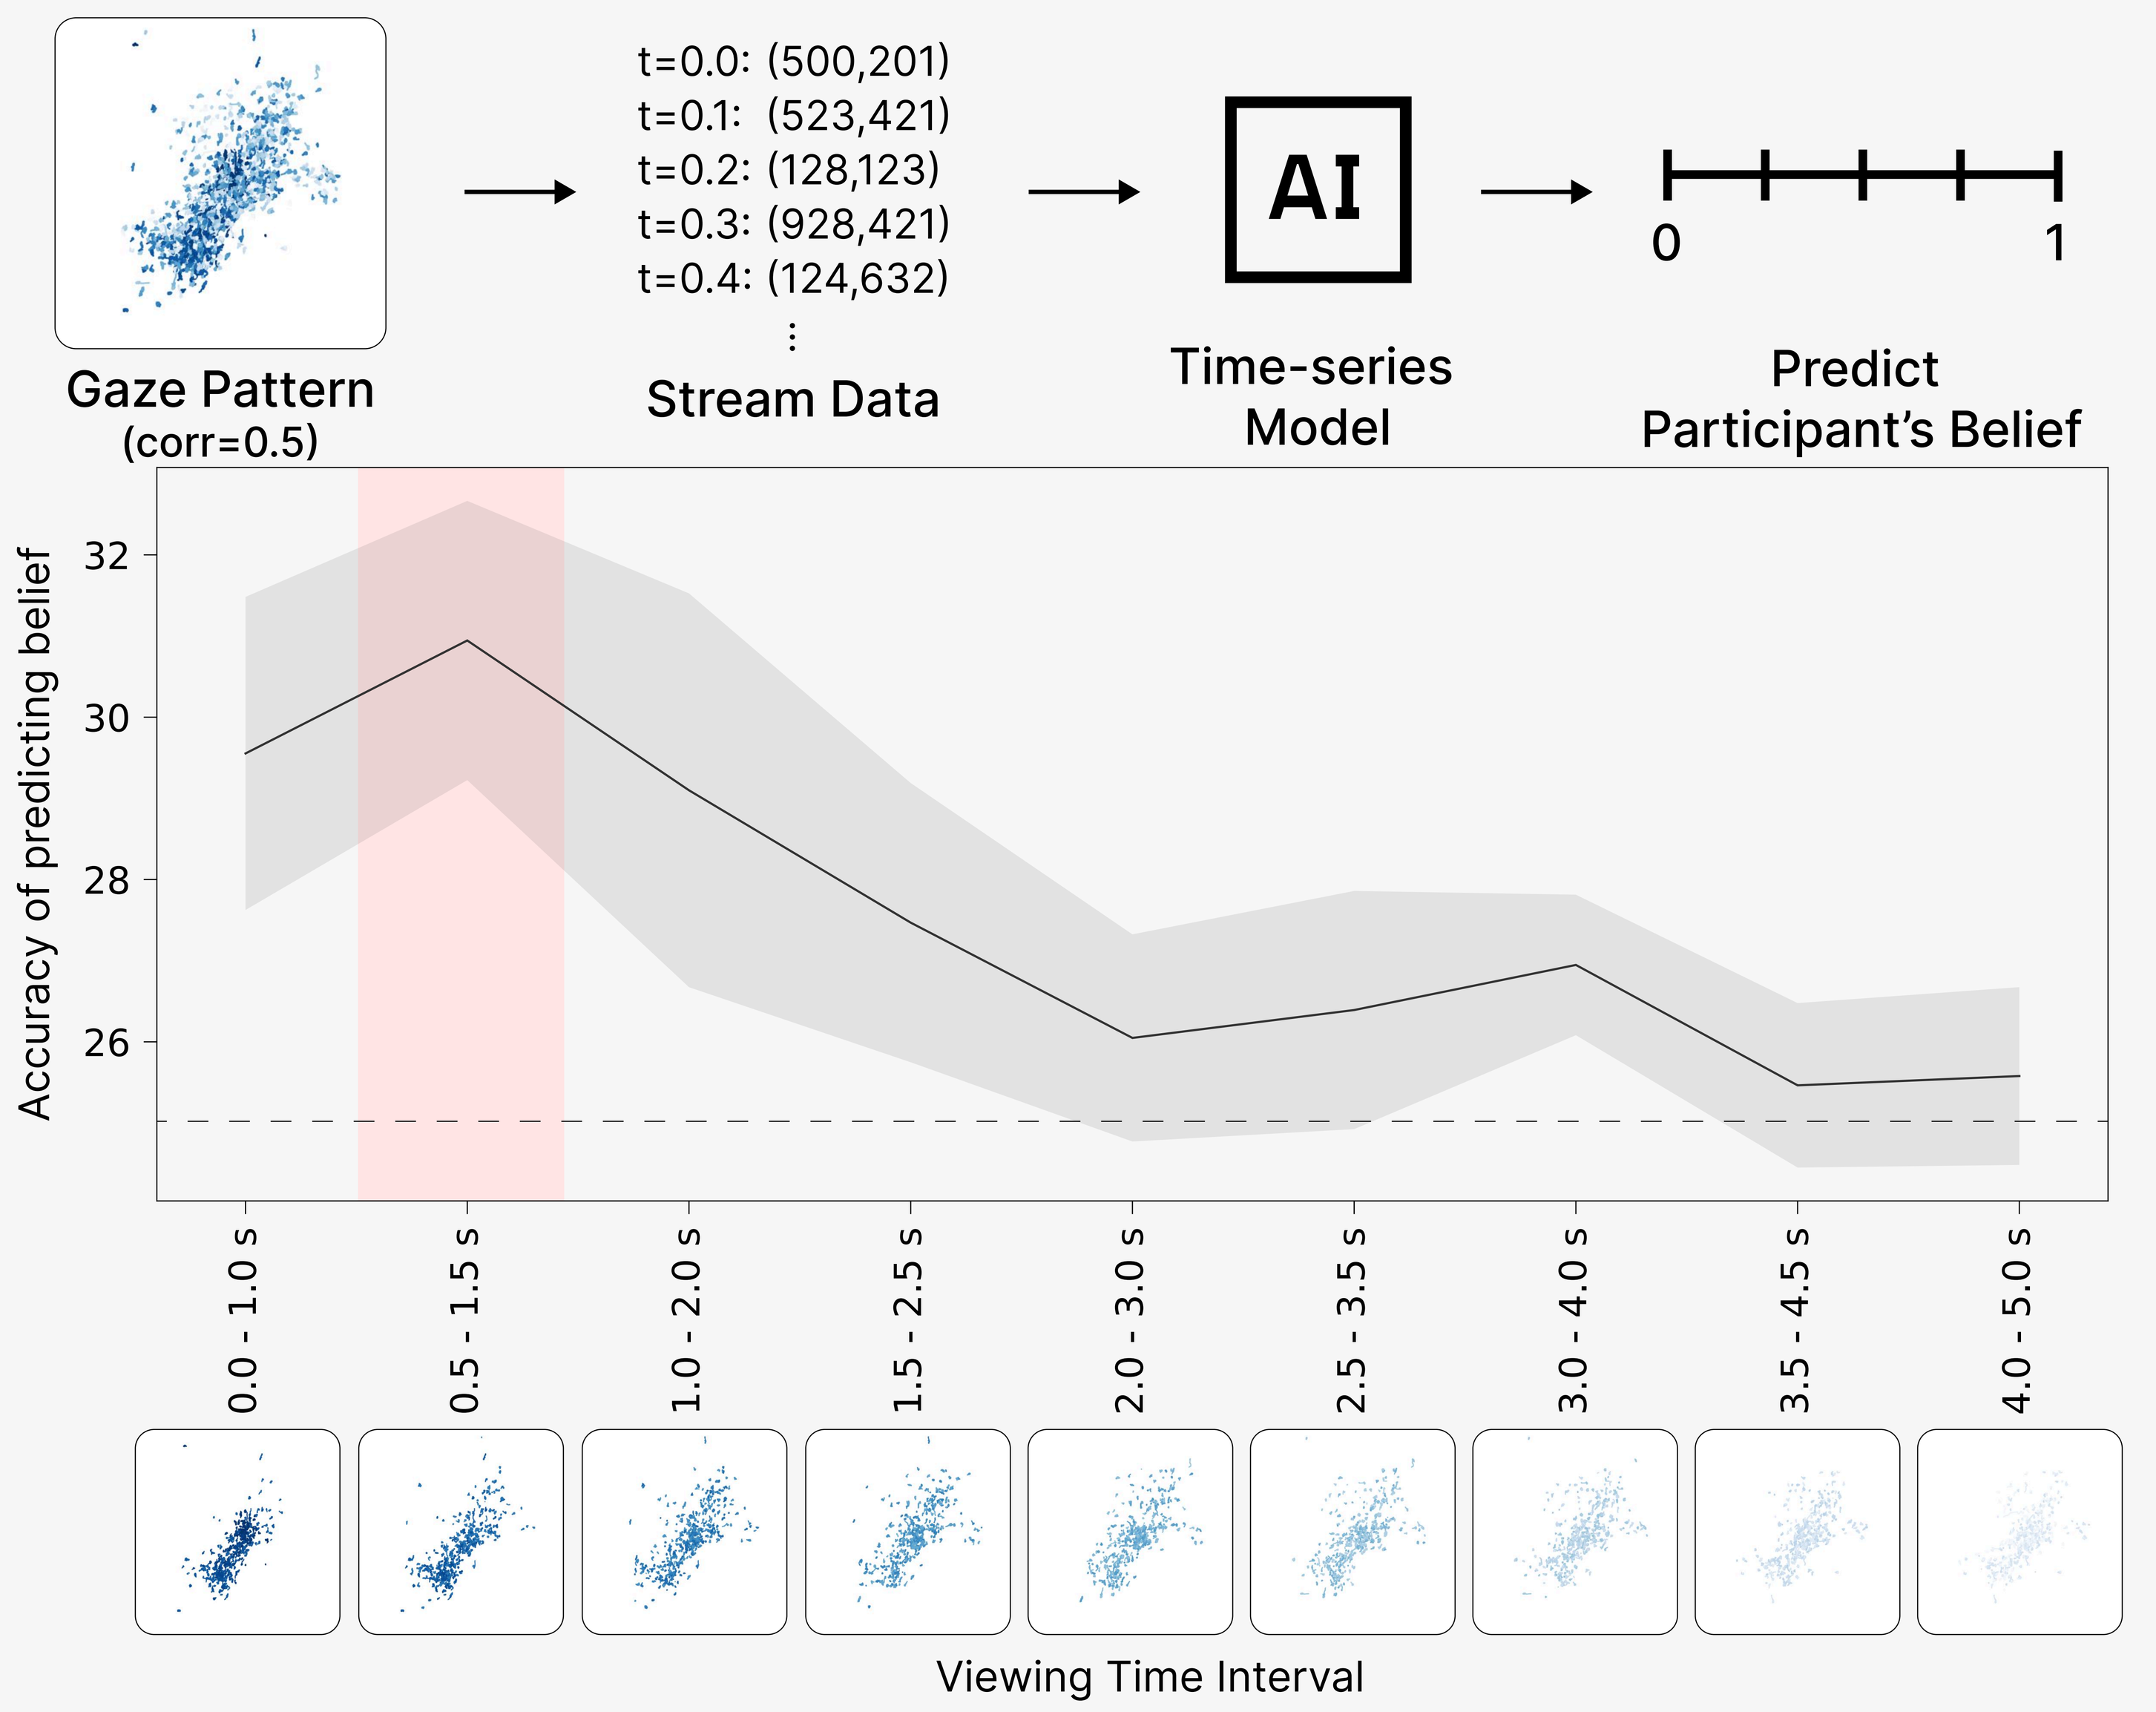
Gaze Pattern
(corr=0.5)
t=0.0: (500,201)
t=0.1: (523,421)
t=0.2: (128,123)
t=0.3: (928,421)
t=0.4: (124,632)
Stream Data
AI
Time-series Model
0
1
Predict Participant's Belief
Accuracy of predicting belief
32
30
28
26
0.0 - 1.0 s
0.5 - 1.5 s
1.0 - 2.0 s
1.5 - 2.5 s
2.0 - 3.0 s
2.5 - 3.5 s
3.0 - 4.0 s
3.5 - 4.5 s
4.0 - 5.0 s
Viewing Time Interval

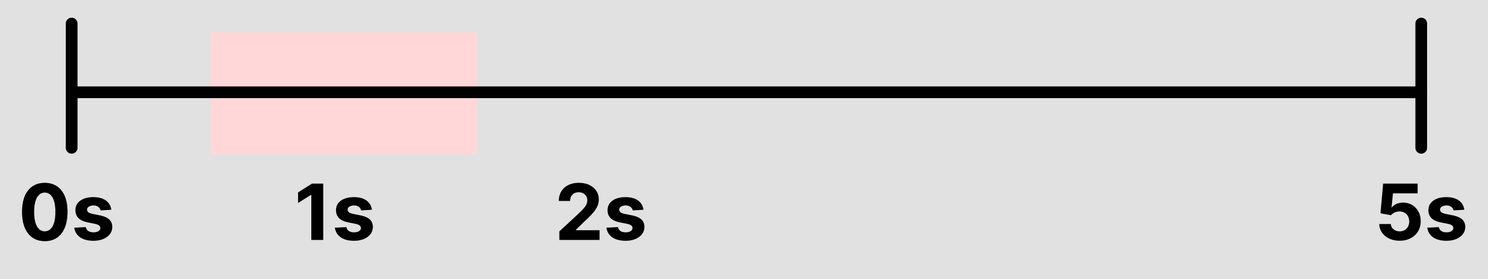
0s
1s
2s
5s

Early Stage Fixations show high influence on **Belief Prediction**

Summary

- We collected **gaze data from 97 participants** using an eye tracker as they performed a **scatterplot correlation estimation task**.
- While people perceive the likability of statements differently, their gaze patterns reveal how strongly they **believe** in those statements.
- Our deep learning experiments show that **early-stage gaze dynamics** contain rich signals for predicting viewers' beliefs.

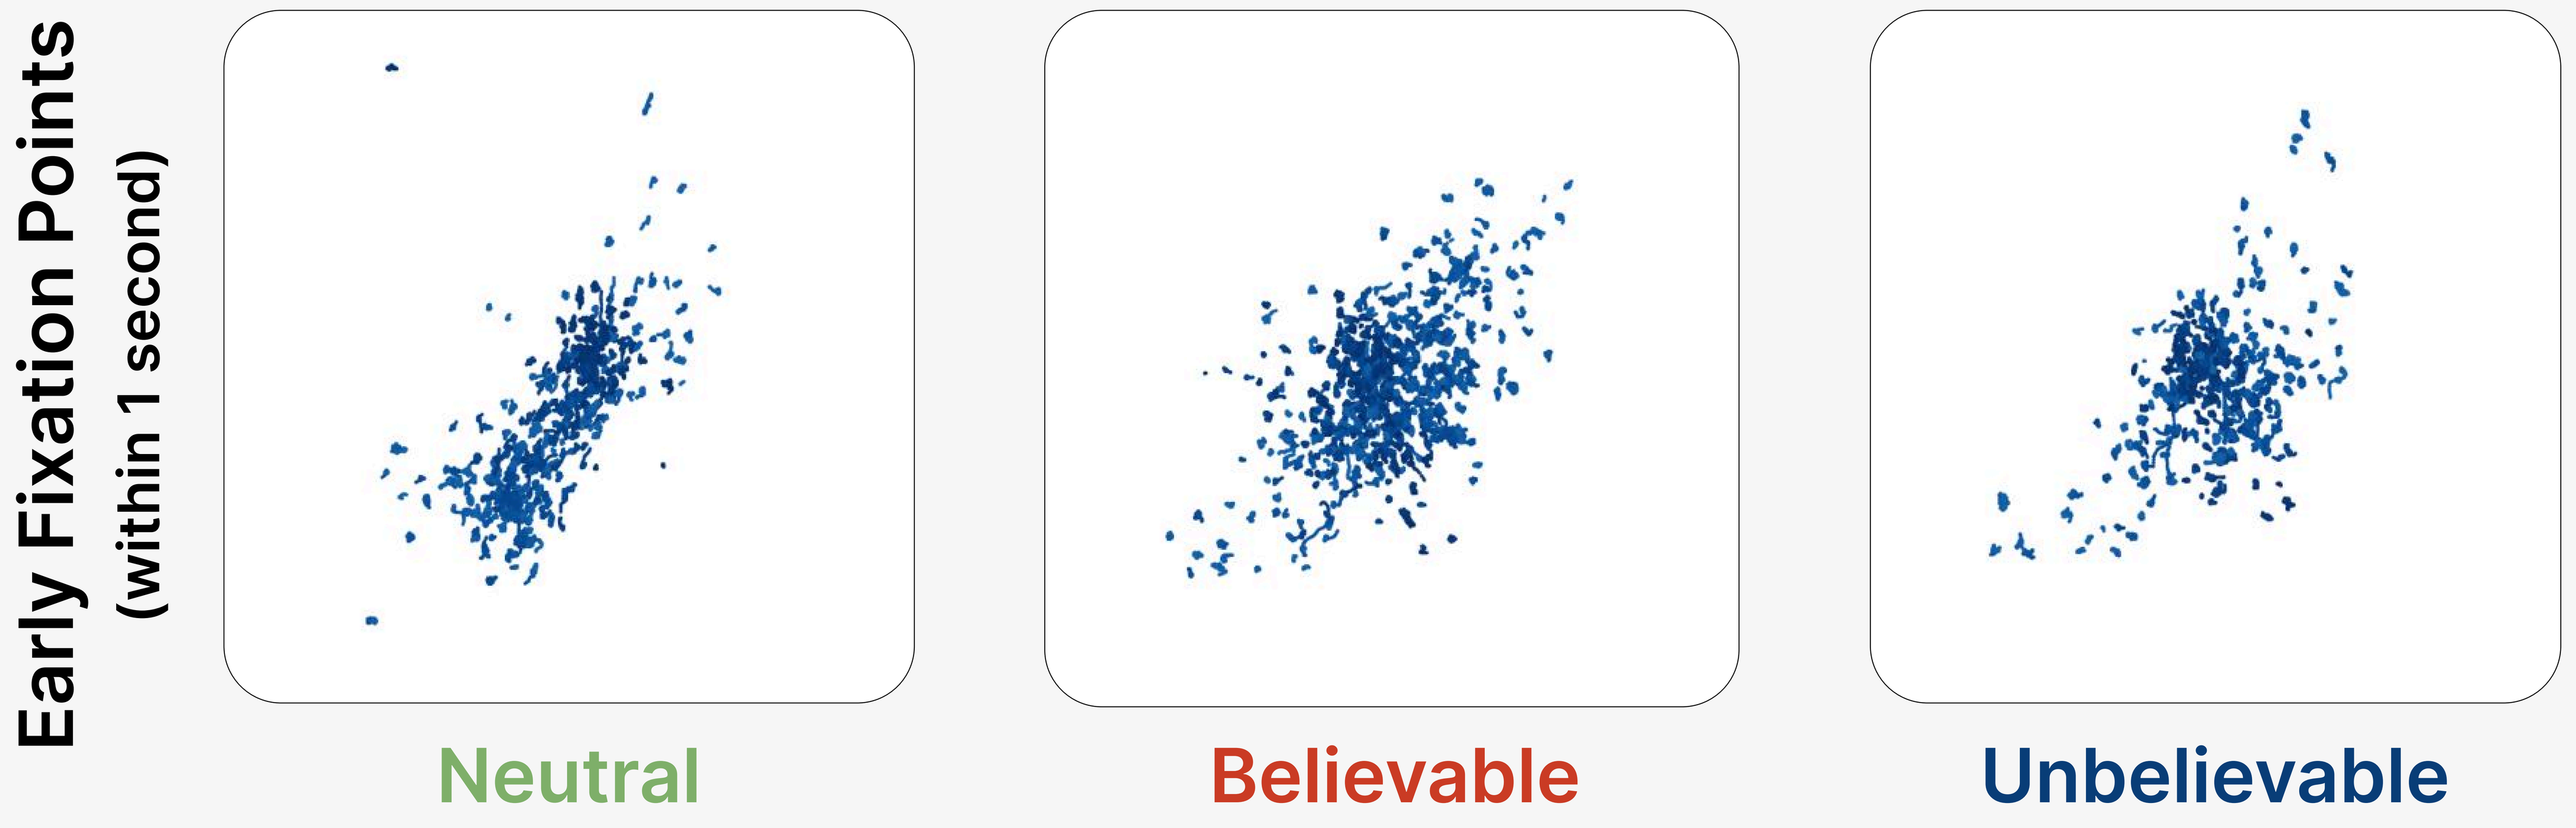
Early Fixation Points
(within 1 second)
Neutral
Believable
Unbelievable

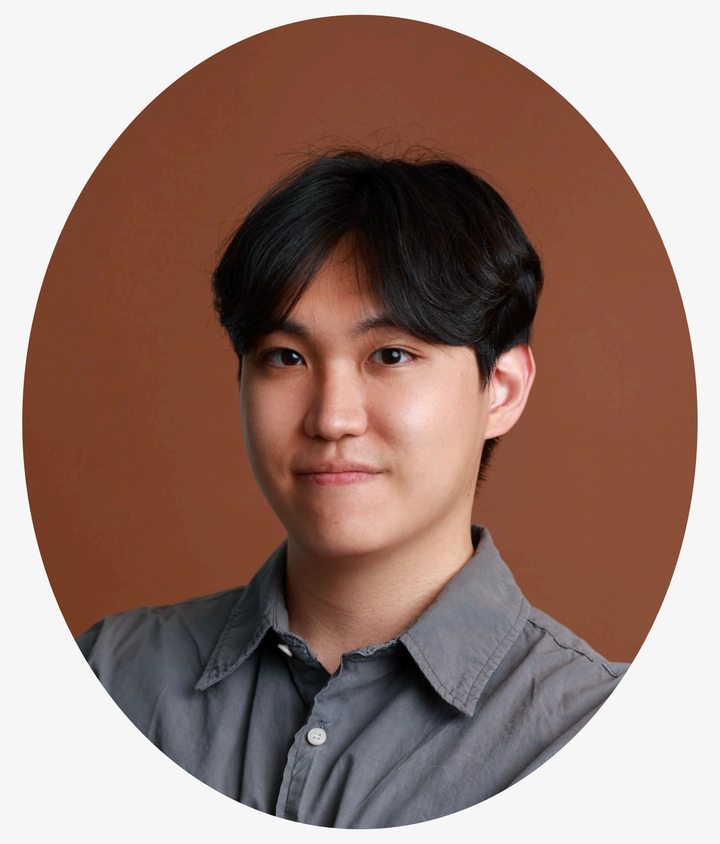

Minsuk Chang

CS PhD Student @ GT

minsuk@gatech.edu
minsukchang.info
@jangsus1

Data Visualization | Graphical Perception | Gaze Tracking